EVALUATION OF THE PROTECTION OF SUNSCREEN PRODUCTS AGAINST LONG WAVELENGTH ULTRAVIOLET A1 AND VISIBLE LIGHT-INDUCED BIOLOGICAL EFFECTS

Nneamaka Ezekwe[1] | Aunna Pourang[1] | Alexis B. Lyons[1] | Shanthi Narla[1] | Amruth Atyam[1] | Shereen Zia[1] | Ben J. Friedman[1] | Iltefat H. Hamzavi[1] | Henry W. Lim[1] | Indermeet Kohli[1,2]

1. Photomedicine and Photobiology Unit, Department of Dermatology, Henry Ford Health System, Detroit, MI 2. College of Physics and Astronomy, Wayne State University, Detroit, MI

INTRODUCTION

- Visible light (VL) has photobiologic effects on the skin such as DNA damage, pigmentation, and erythema[1].
- Long wavelength Ultraviolet A1 (UVA1) +VL has been shown to create synergistic, effects of erythema in light-skinned individuals and both erythema and pigmentation in dark-skinned individuals[2].
- There is a need for photoprotection against broader wavelengths.

STUDY OBJECTIVE:

To determine the protective effect of formulated topical products on long wavelength UVA1 and visible light (VL+UVA1) biologic effects.

METHODS & ASSESSMENTS

METHODOLOGY:

- Completed study with n=12 (10 female, 2 male) participants, of which 8 had Skin Phototypes (SPT) III, and 4 had SPT IV.
- Visit 1 (Day 0): Baseline assessment
 - Irradiation with VL+UVA1 dose: 320 J/cm2 after 30 minutes of topical product application
 - Immediate pigment darkening (IPD) and erythema assessments
- Visit 2 (Day 1): Persistence pigment darkening (PPD) assessment
- Visit 3 (Day 7): Delayed Tanning (DT) assessment
- Visit 4 (Day 14): Persistence of Delayed Tanning (DT) Assessment

ASSESSMENTS:

- Subjective measurements: Investigator's Global Assessment (IGA) score for erythema and pigmentation and Photography
- Objective measurements: Colorimetry (Delta a*: Erythema; Delta L*: Pigmentation)

TABLE 1 Composition of the formulations tested.

Product	SPF	Broad spectrum	UV filters	Antioxidants	Iron oxides
Product A	60	Yes	Avobenzone 3%	Tocopherol[a] and Cassia alata leaf extract[b]	No
			Homosalate 10%		
			Octisalate 5%		
			Octocrylene 7%		
Product B	50	Yes	Homosalate 15%	Tocopherol and Cassia alata leaf extract	Iron oxides 4%
			Octisalate 5%		
			Octocrylene 7%		
			Zinc oxide 12%		
Product C	50	Yes	Homosalate 15%	Tocopherol and Cassia alata leaf extract	No
			Octisalate 5%		
			Octocrylene 7%		
			Zinc oxide 12%		
Product D	50	Yes	Titanium dioxide 11% (micronized)	Tocopherol and Cassia alata leaf extract	Iron oxides 1%

Note: Further details of formulations are considered proprietary information.
[a]Concentration in product A slightly higher (approximately 2.5×) than products B, C, and D.
[b]Same concentration in all four products.

TABLE 2 Erythema and pigmentation clinical scoring scales used for IGA.

Score	Description of erythema
0	Clear of erythema
1	Almost clear of erythema
2	Mild, but noticeable erythema
3	Moderate erythema (pink in quality), no sharp borders
4	Severe erythema (dark pink in quality), sharp borders
5	Very severe erythema (very dark pink, almost red in quality)

Score	Description of pigmentation (tanning)
0	Clear of hyperpigmentation
1	Almost clear of hyperpigmentation
2	Mild, but noticeable hyperpigmentation
3	Moderate hyperpigmentation (medium brown in quality)
4	Severe hyperpigmentation (dark brown in quality)
5	Very severe hyperpigmentation (very dark brown, almost black in quality)

RESULTS

- Sites treated with products C and D had a statistically significant decrease (P<0.05) in IGA erythema scores immediately after irradiation compared to the control (irradiated, untreated site).
- Sites treated with product D reached significance per colorimetry a* (erythema) parameter.
- IGA pigmentation scores were statistically significantly lower for sites B and D on day 0, all sites (A, B, C, D) on day 7, and site D for day 14 compared to control.
- Colorimetric L* (pigmentation data showed a significant decrease in pigmentation for sites B and D at day 0 with a similar downward trend at sites B and D on day 14.

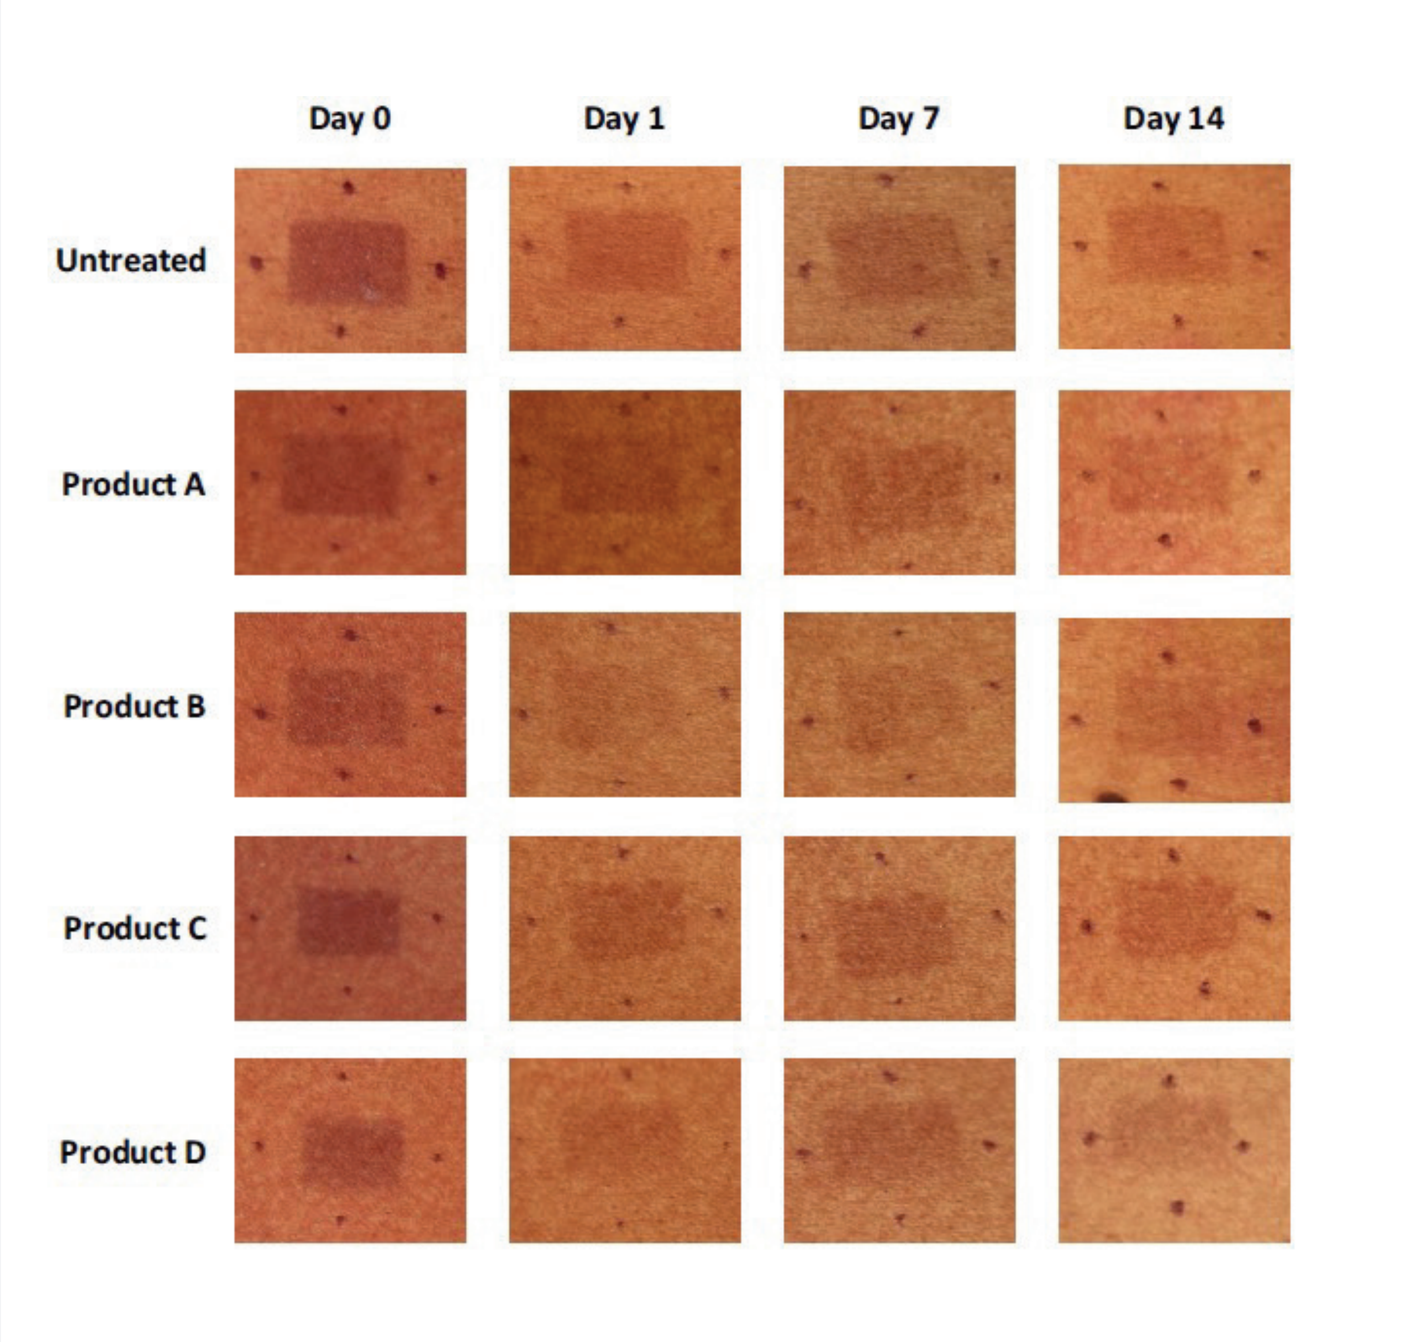
Day 0
Day 1
Day 7
Day 14
Untreated
Product A
Product B
Product C
Product D

Figure 1: Representative photographs of one participant at all time points.

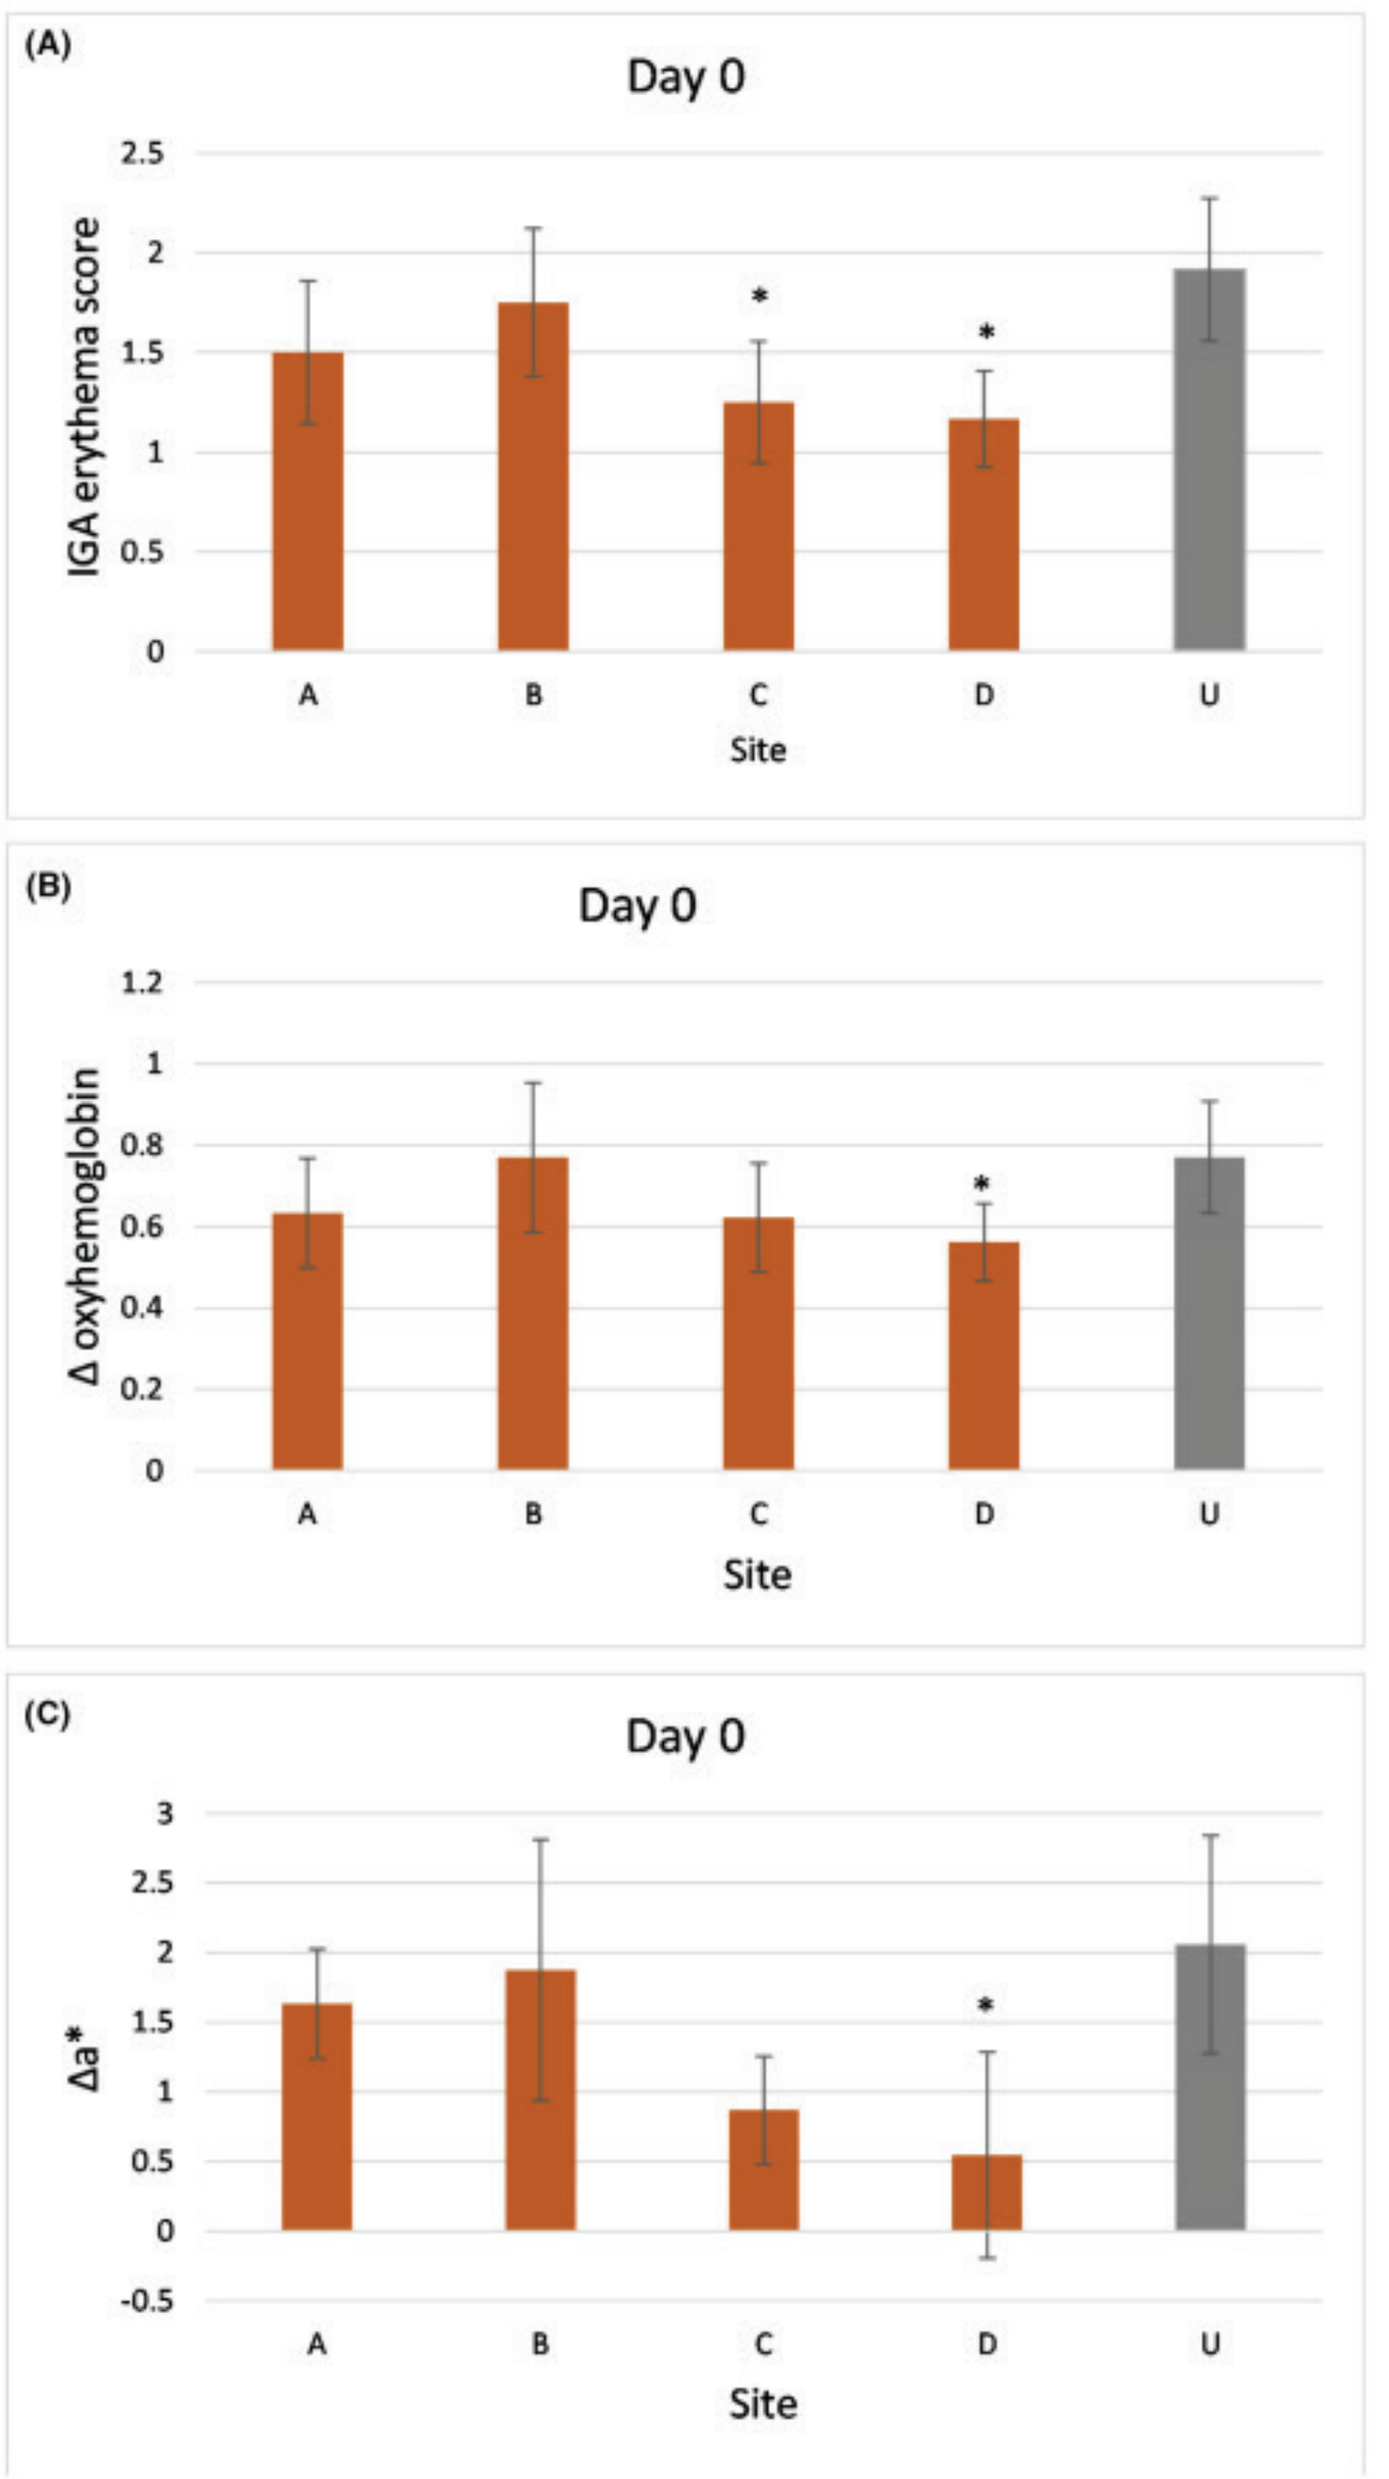
(A)
Day 0
IGA erythema score
Site
(B)
Day 0
Δ oxyhemoglobin
Site
(C)
Day 0
Δa*
Site

Figure 2: Erythema at Day 0 according to (A) IGA scores, (B) DRS Δ oxyhemoglobin, higher values indicate more erythema (C) colorimetry Δa*, higher values indicate more erythema. Data are mean and SEM. Day 0 refers to immediately after irradiation. p < 0.05

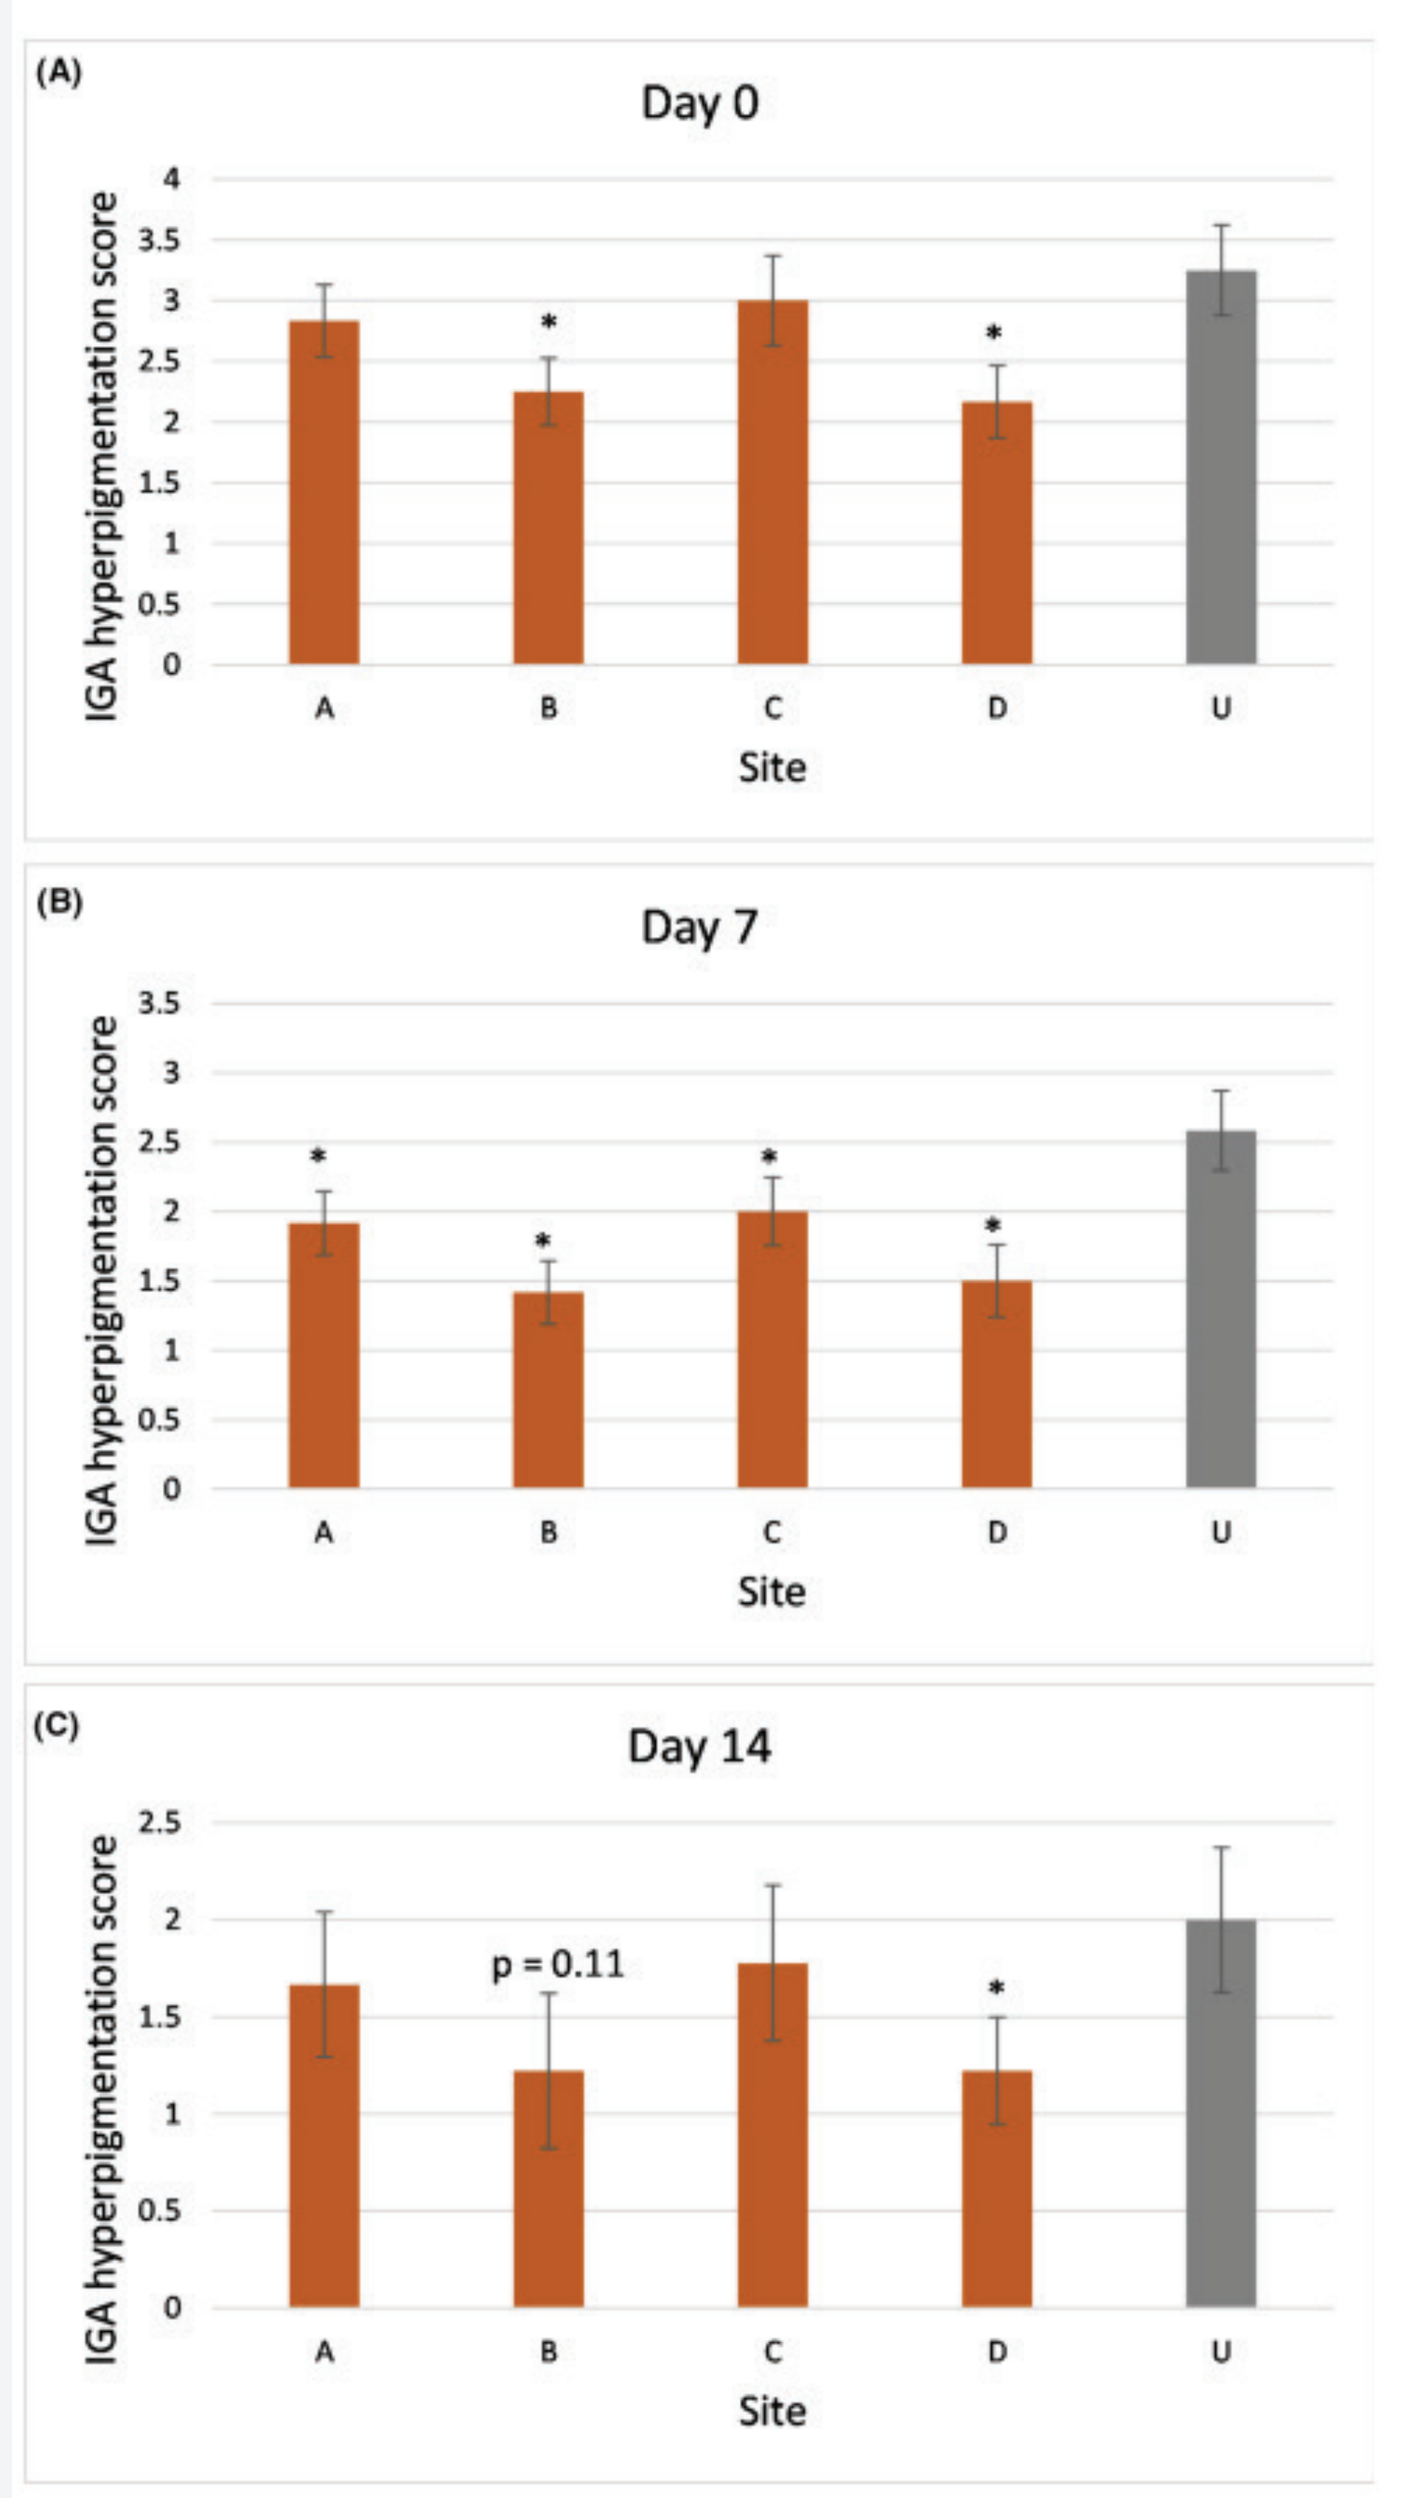
(A)
Day 0
IGA hyperpigmentation score
Site
(B)
Day 7
IGA hyperpigmentation score
Site
(C)
Day 14
p = 0.11
IGA hyperpigmentation score
Site

Figure 3: Clinical hyperpigmentation based on IGA assessment at Days 0 (A), 7 (B), and 14 (C). Day 0 refers to immediately after irradiation. Data are mean and SEM. p < 0.05

CONCLUSIONS

- ✓ TINTED SUNSCREEN WITH INORGANIC FILTERS PROVIDED SUPERIOR PROTECTION AGAINST VL+UVA1-INDUCED IMMEDIATE ERYTHEMA AND DELAYED PIGMENTATION.
- ✓ UNTINTED PRODUCT WITH ZINC OXIDE AND ORGANIC FILTERS (PRODUCT C), TINTED PRODUCT WITH TITANIUM DIOXIDE (D), SIGNIFICANTLY SUPPRESSED UVA1 and VL-INDUCED ERYTHEMA ON DAY 0.
- ✓ ALL PRODUCTS TESTED SUPPRESSED PIGMENTATION; HOWEVER, THE MOST EFFICIENT SUPPRESSION WAS BY TINTED PRODUCTS (B AND D).
- ✓ FURTHER RESEARCH IS NEEDED TO EVALUATE IF IRON OXIDE CONTENT IN TINTED PRODUCTS CORRELATES WITH SUPERIOR PROTECTION.

References:

1. Narla S, Kohli I, Hamzavi IH, Lim HW. Visible light in photodermatology. Photochem Photobiol Sci. 2020;19(1):99- 104. doi:10.1039/c9pp00425d 2. Kohli I, Chaowattanapanit S, Mohammad TF, et al. Synergistic effects of long-wavelength ultraviolet A1 and visible light on pigmentation and erythema. Br J Dermatol. 2018;178(5):1173-1180. doi:10.1111/bjd.15940

This work was supported by La Roche-Posay Laboratoire Dermatologique.

EFFICACY OF A 2-MNG-CONTAINING SERUM AND SUNSCREEN REGIMEN ON IMPROVING FACIAL-DYSCHROMIA IN SKIN OF COLOR WOMEN

Hawasatu Dumbuya[1], Stephen Lynch[2], Katharine Podimatis[1]

Disclosure Information: [1]*La Roche-Posay Laboratoire Dermatologique, L'Oreal USA, New York, NY;* [2] *L'Oréal USA Research and Innovation, Clark, NJ;* All authors are employees of L'Oréal USA.

INTRODUCTION

Due to higher levels of melanin in their skin, SOCs are more susceptible to experiencing dyschromia (hyper- and hypo-pigmentation), leading to uneven skin tone, ashy skin and blotchiness.[1,2] Despite skin discoloration being considered as one of the top dermatological concerns for SOCs, they remain under-represented in clinical trials focused on skin-aging prevention and photoprotection.[3]

STUDY OBJECTIVE:

To evaluate the efficacy of an innovative serum and sunscreen regimen containing 2-MNG, an ingredient that quenches melanin precursors, on improving facial dyschromia in women of color following 12 weeks of usage.

DESIGN & METHODS

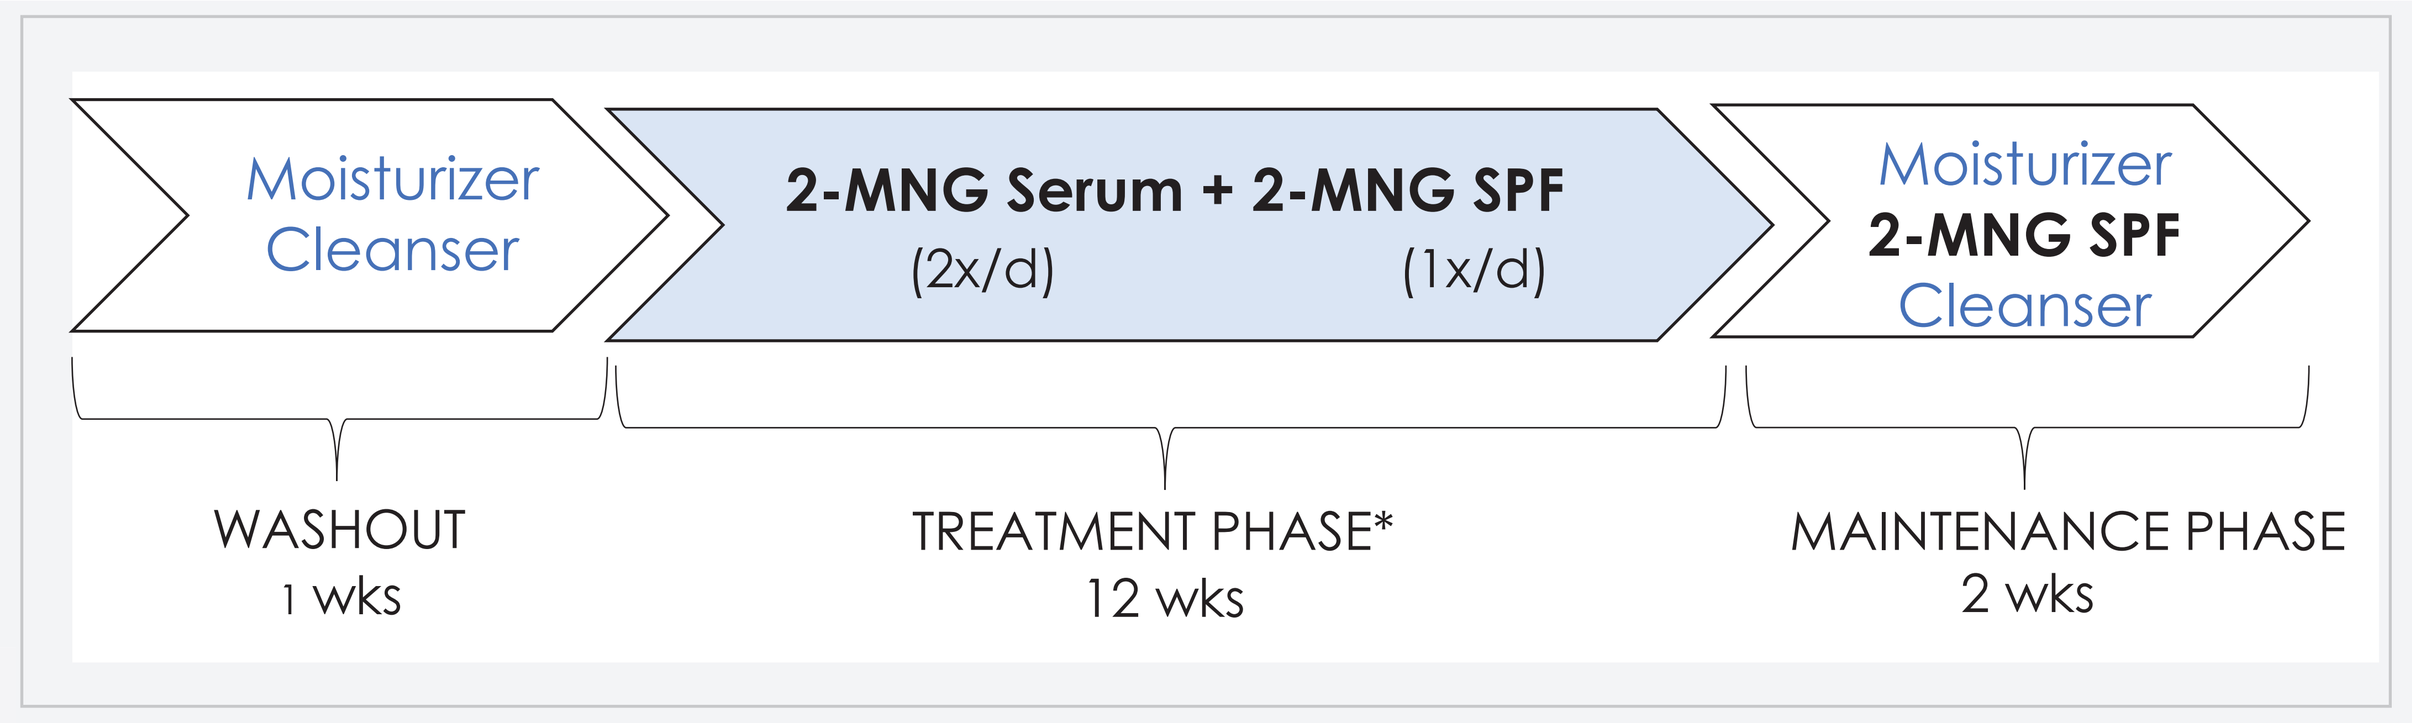
Moisturizer
Cleanser
2-MNG Serum + 2-MNG SPF
(2x/d)
(1x/d)
Moisturizer
2-MNG SPF
Cleanser
WASHOUT
1 wks
TREATMENT PHASE*
12 wks
MAINTENANCE PHASE
2 wks

DEMOGRAPHICS:

- Monocenter, double-blinded study
- Completed study with n=60 female patients ages 25-70 yo
- From diverse racial/ethnic backgrounds with skin phototype IV-VI
- Presenting with mild to moderate uneven skin tone, hyperpigmentation, and skin roughness

TREATMENT ROUTINE:

After completing a 1-week washout period, all subjects started using a 2-MNG -containing product regimen, consisting of a serum (applied morning and evening) and a sunscreen SPF 30 (applied 15min before sun exposure) for 12 weeks. All subjects were given the same moisturizer to use when needed at night (after serum application) and instructed to use their own cleanser throughout study.

EVALUATIONS (BASELINE, WEEK 2, 4, 8 & 12):

Evaluations included clinical grading, quality-of-life questionnaires, plus clinical imaging at indicated time points.

RESULTS

FIGURE 1: 2-MNG-CONTAINING PRODUCT REGIMEN (SERUM & SPF30) IMPROVES FACIAL DYSCRHOMIA IN WOMEN OF COLOR. (A & B) Clinical assessments by dermatologist showed significant improvement in dyschromia, skin tone evenness, radiance, plus other skin discoloration-related endpoints in all subjects by Week 12. ** denotes p<0.05 statistically significant different vs. Baseline.*

IMPROVEMENT OF FACIAL DISCRHORMA-RELATED ENDPOINTS

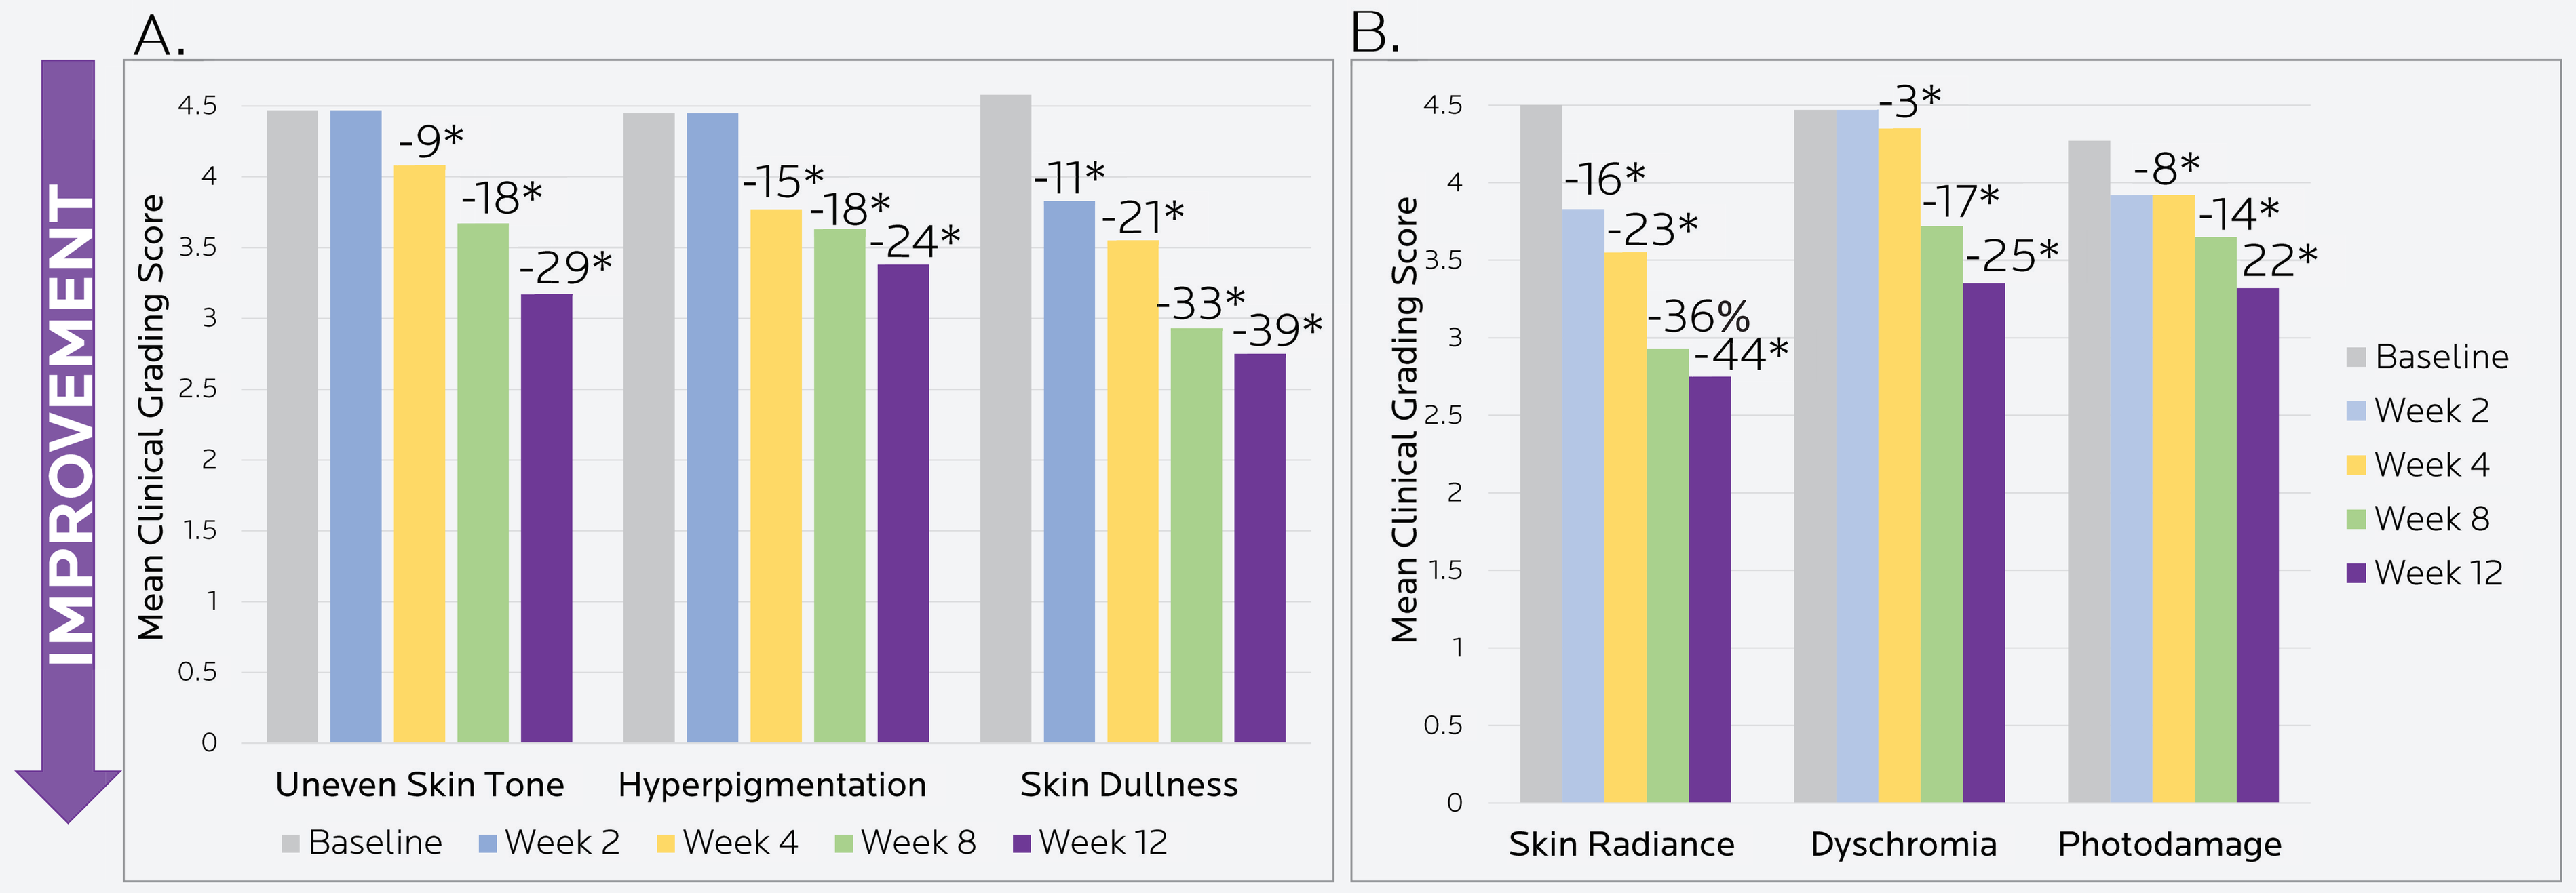
A.
B.
IMPROVEMENT
Mean Clinical Grading Score
-9*
-18*
-29*
-15*
-18*
-24*
-11*
-21*
-33*
-39*
Uneven Skin Tone
Hyperpigmentation
Skin Dullness
-16*
-23*
-36%
-44*
-3*
-17*
-25*
-8*
-14*
22*
Skin Radiance
Dyschromia
Photodamage
Baseline
Week 2
Week 4
Week 8
Week 12

RESULTS CONT.

FIGURE 2: 2-MNG-CONTAINING PRODUCT REGIMEN (SERUM & SPF30) IMPROVES SKIN TEXTURE IN WOMEN OF COLOR. (A & B) Clinical assessments by dermatologist showed significant improvement in overall appearance and skin texture-related endpoints in all subjects by Week 12. ** denotes p<0.05 statistically significant different vs. Baseline.*

IMPROVEMENT OF SKIN TEXTURE & OVERALL APPEARANCE

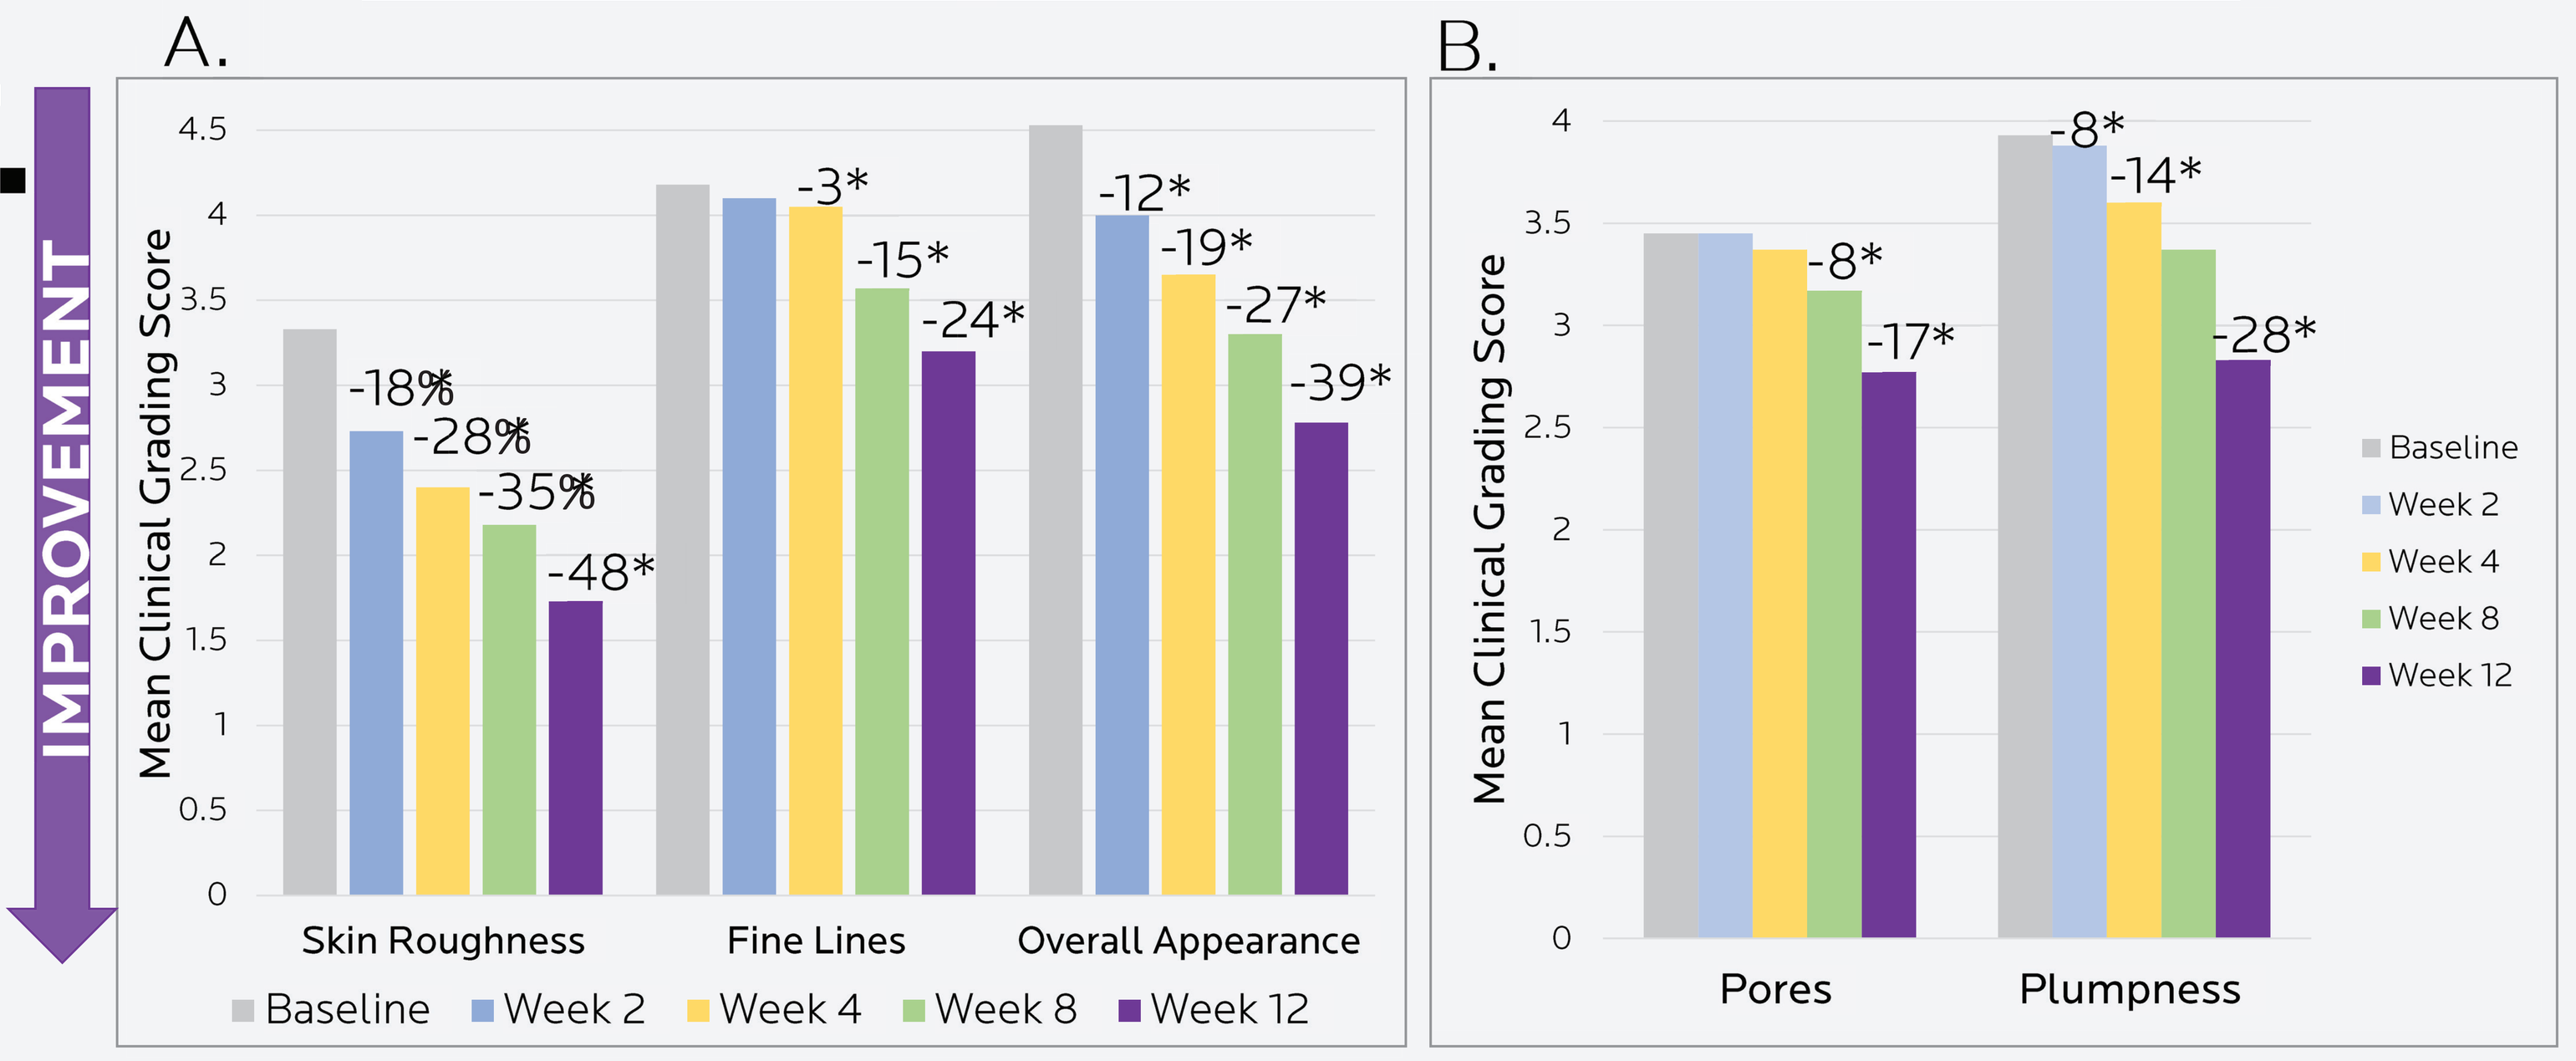
A.
B.
IMPROVEMENT
Mean Clinical Grading Score
-18%
-28%
-35%
-48*
-3*
-15*
-24*
-12*
-19*
-27*
-39*
Skin Roughness
Fine Lines
Overall Appearance
-8*
-17*
-8*
-14*
-28*
Pores
Plumpness
Baseline
Week 2
Week 4
Week 8
Week 12

FIGURE 3: 2-MNG-CONTAINING PRODUCT REGIMEN (SERUM & SPF30) EFFICACY WAS PERCEIVED BY PATIENTS AND WAS CONSISTENT WITH IMPROVED QUALITY OF LIFE OF. Via self-assessments questionnaires, female patients of color perceived (A) significant improvement of skin brightness, dark spots visibility and skin tone evenness, (B) plus significant improvement in overall quality-of-life overtime after using product regimen. ** denotes p<0.05 statistically significant different vs. Baseline.*

A. PERCEIVED PRODUCT REGIMEN EFFICACY BY PATIENTS

B. PERCEIVED IMPROVEMENT OF QUALITY OF LIFE BY PATIENTS

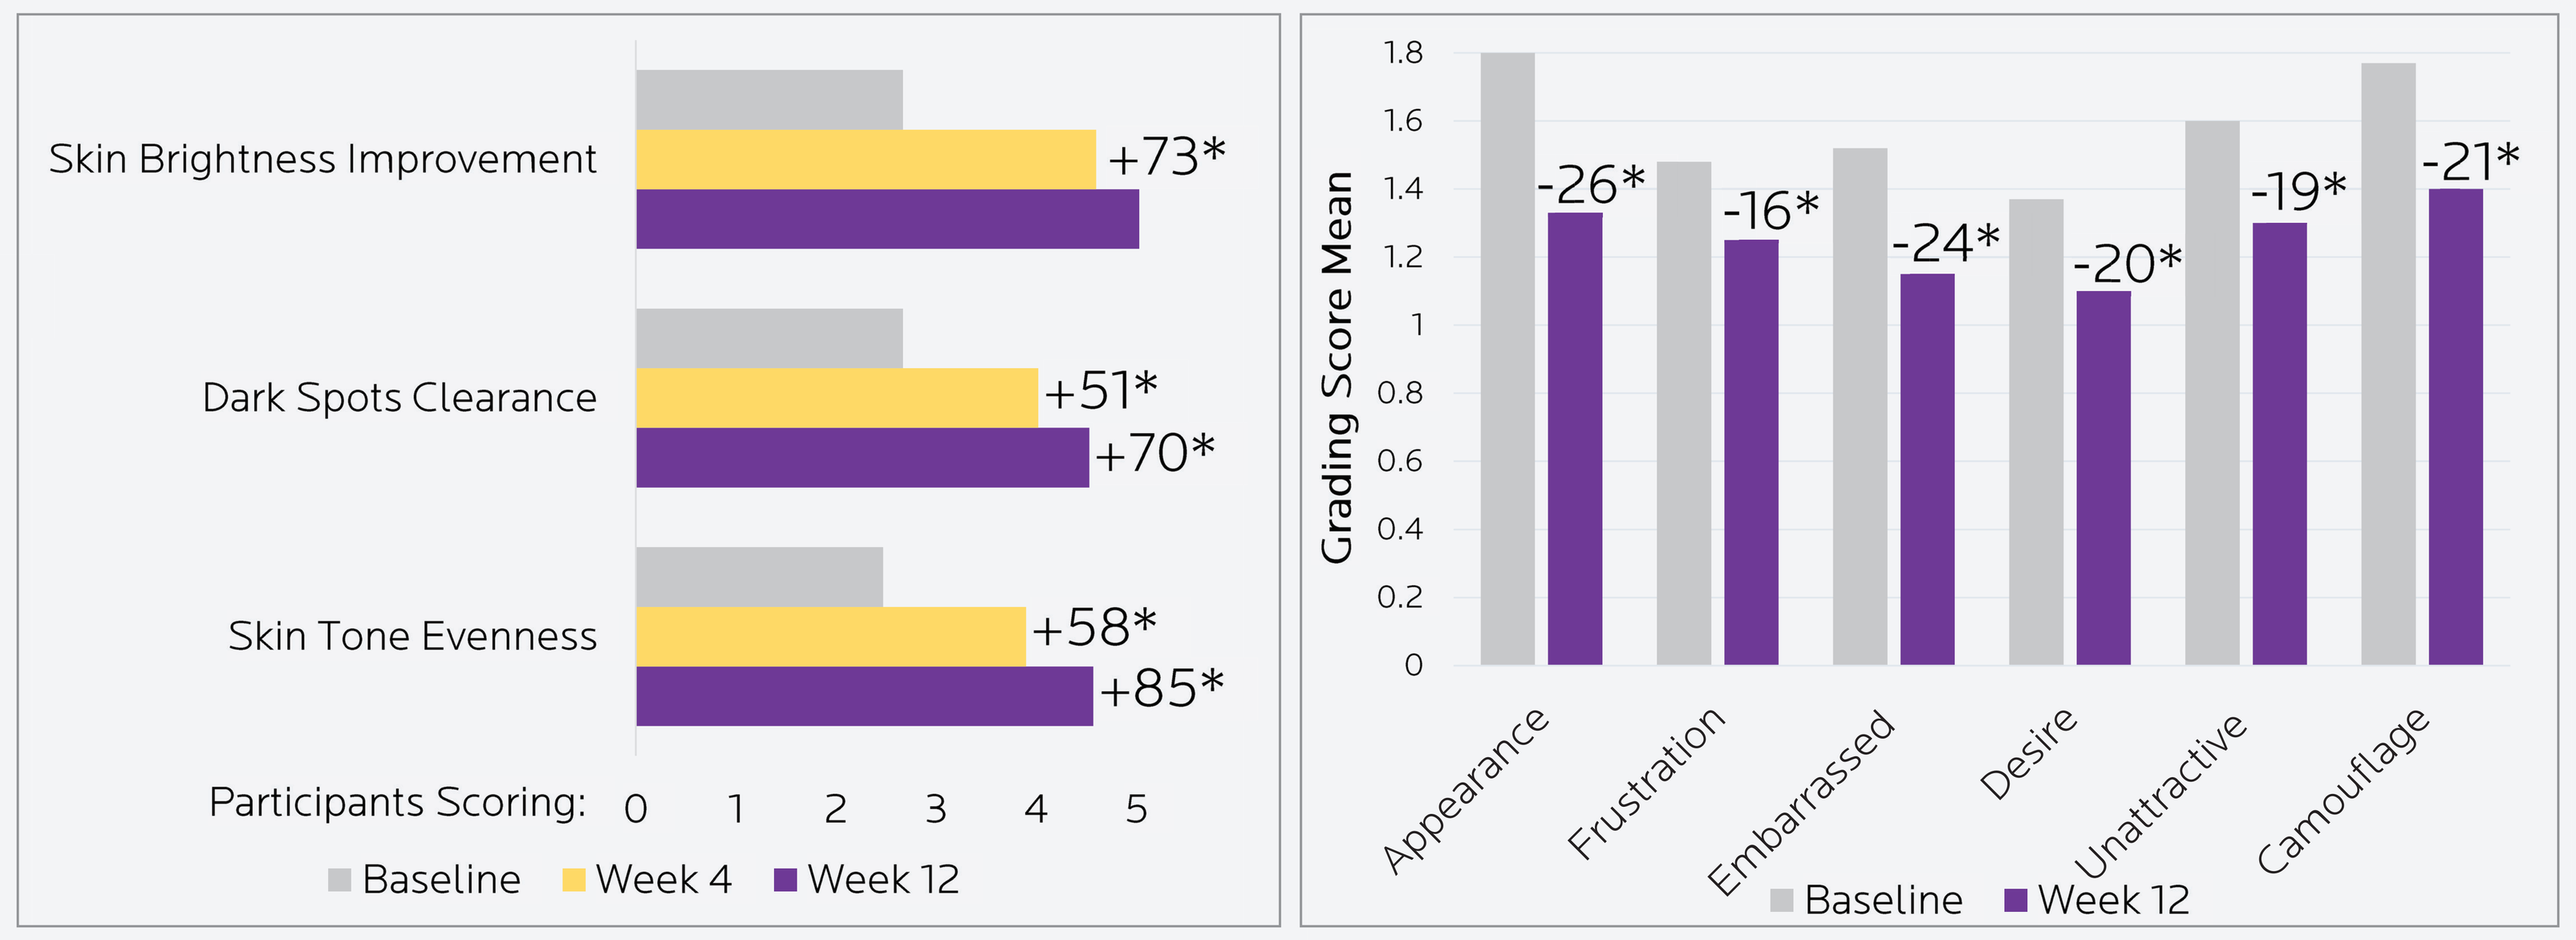
Skin Brightness Improvement
+73*
Dark Spots Clearance
+51*
+70*
Skin Tone Evenness
+58*
+85*
Participants Scoring: 0 1 2 3 4 5
Baseline
Week 4
Week 12
Grading Score Mean
-26*
-16*
-24*
-20*
-19*
-21*
Appearance
Frustration
Embarrassed
Desire
Unattractive
Camouflage
Baseline
Week 12

FIGURE 4: REPRESENTATIVE IMAGES OF AVERAGE RESPONDERS FOLLOWING 12 WEEKS OF 2-MNG-CONTAINING PRODUCT REGIMEN (SERUM & SPF30). (A&B) Overall significant improvement in facial dyschromia and skin texture appearance overtime in women of color. Images were captured using VISIA imaging system.

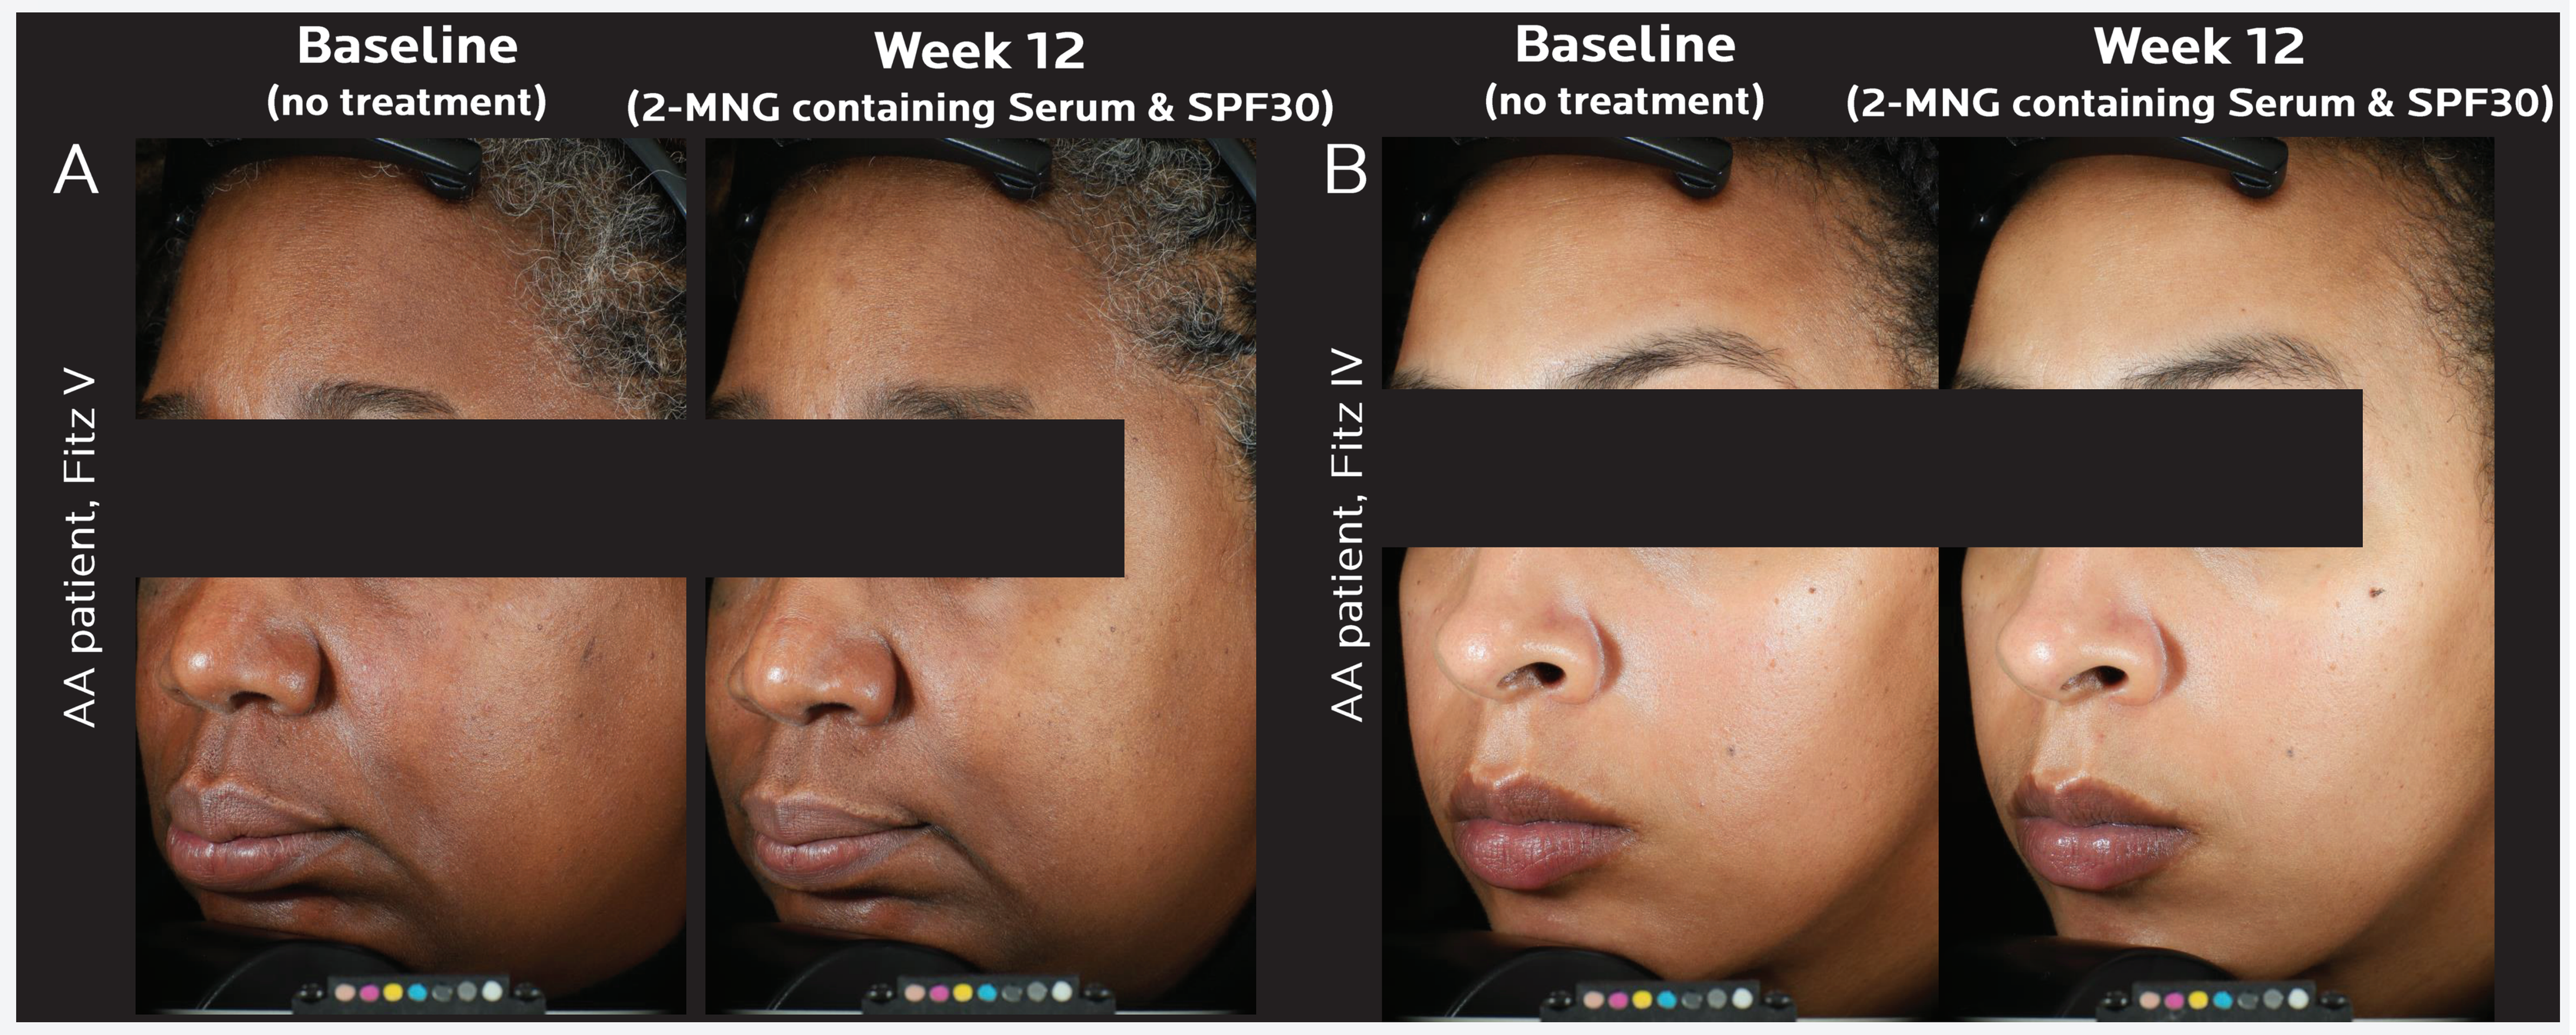
Baseline
(no treatment)
Week 12
(2-MNG containing Serum & SPF30)
Baseline
(no treatment)
Week 12
(2-MNG containing Serum & SPF30)
A
AA patient, Fitz V
B
AA patient, Fitz IV

CONCLUSIONS

- ✓ OUR RESTULTS DEMONSTRATE THAT A 2-MNG CONTAINING PRODUCT REGIMEN, CONSISTING OF A SERUM AND A SUNSCREEN SPF30, CAN EFFECTIVELY IMPROVE OVERALL SKIN TONE AND QUALITY IN PATIENTS OF COLOR WITH FACIAL DYSCHROMIA-RELATED CONCERNS.
- ✓ THE IMPROVEMENT IN OVERALL QUALITY-OF-LIFE EXPERIENCED BY CLINICAL PARTICIPANTS AFTER USING PRODUCT REGIMEN OVERTIME MAY HELP SUPPORT CLINICIANS ON SKIN_AGING PREVENTION AND PHOTOPROTECTION STRATEGES TO CONSIDER FOR ALL PATIENTS, PARTICULARLY FOR PATIENTS OF COLOR.

References:
1. Venkatesh S, Maymone MBC, Vashi NA, et al. *Aging in skin of color.* Clin Dermatol. 2019 Jul-Aug;37(4):351-357.
2. Campiche R, Trevisan S, Séroul P, et al. Appearance of aging signs in differently pigmented facial skin by a novel imaging system. *J Cosmet Dermatol. 2019 Apr;18(2):614-627.*
3. Callender VD, Harvey VM, Hartman CL, et al. Do Women with Skin of Color Think They Are Well Represented in Skin Aging Prevention Information? *J Clin Aesthet Dermatol. 2024 Apr;17(4):18-22.*

Acknowledgement: We thank Emma DiMartini for her valuable contributions on the poster design

This work was supported by La Roche-Posay Laboratoire Dermatologique US

Improved Compliance of Daily Sunscreen Application Through a Simple, Daily, Goal-Setting Reminder

Ethan D. Paliwoda MBA,[1] Elle Waske,[2] Steven Q. Wang MD[3]

[1] Albany Medical College, Albany, NY; [2] University of California Los Angeles, Los Angeles, CA; [3] Hoag Memorial Hospital Presbyterian, Irvine, CA

Abstract

Introduction:

Sunscreen is crucial in preventing sunburn, skin cancers, and premature aging, yet consistent usage remains a challenge despite widespread awareness efforts.

Methods:

In a prospective and randomized study, participants were assigned to use previously weighted sunscreen alone or with a calendar-based tracking system. Eligible individuals were randomly assigned, educated on sunscreen benefits, and provided with a two-month supply of SPF-30 sunscreen. The calendar group was instructed to mark each day of sunscreen application and told to not “break the chain.” The volume remaining in the distributed sunscreen was measured at follow-up appointments 6 weeks later.

Results:

Among 53 participants, the calendar group demonstrated significantly higher sunscreen utilization compared to the control group. The control group used 15.96% of the sunscreen, the calendar group used 29.92% of the sunscreen, resulting in an 23.34% of more usage of sunscreen in the calendar group (p<0.001). There was a significant difference in volume of sunscreen used between the study arms, with the calendar group using 0.84 ounces more than the control group (p<0.001)

Conclusion:

Integrating practical tools like calendars alongside education may effectively reinforce sunscreen application habits. These findings highlight the potential of such interventions in promoting sustained sunscreen adherence and reducing sun-related skin damage. Further research is needed to validate these results on a larger scale, but they offer valuable insights for future public health initiatives aimed at improving sunscreen compliance.

Introduction

Sunscreen mediates preventing sunburn, skin cancers, and premature aging caused by sun exposure. Over time, dermatologists and academic institutions as well as commercial and non-profit organizations have consistently worked to educate the public about the importance of proactive sunscreen use. Despite widespread awareness of its benefits, many individuals still fail to incorporate sunscreen into their daily routine for various reasons. Further research into interventions to increase compliance rates is needed.[1-4]

Objective

To demonstrate the feasibility of improving compliance with the daily application of sunscreen by performing a simple, daily, goal-setting reminder.

Methods

This randomized, parallel-group study design involved participants randomly assigned to either Sunscreen Group (sunscreen instruction only) or Calendar Group (sunscreen instruction + calendar). All participants received education on daily sunscreen benefits, completed a baseline questionnaire on skin cancer history and photoprotection motivations, and received a weighted 2-month supply of of broad spectrum SPF-30 sunscreen (La Roche Posay Product). Calendar Group additionally received a calendar to track daily sunscreen use, with instructions to mark each day sunscreen was applied and aim to maintain a continuous "chain" of marks without breaks. Participants were instructed to return after 6 weeks for sunscreen weighing and questionnaire completion. The primary outcome measured the difference in remaining sunscreen volumes between groups. Regression modeling assessed group differences, considering participant characteristics. Secondary outcomes included counts of marked calendar dates, evaluated for group differences and correlations with sunscreen weight differences. IRB approval preceded the study start.

Results

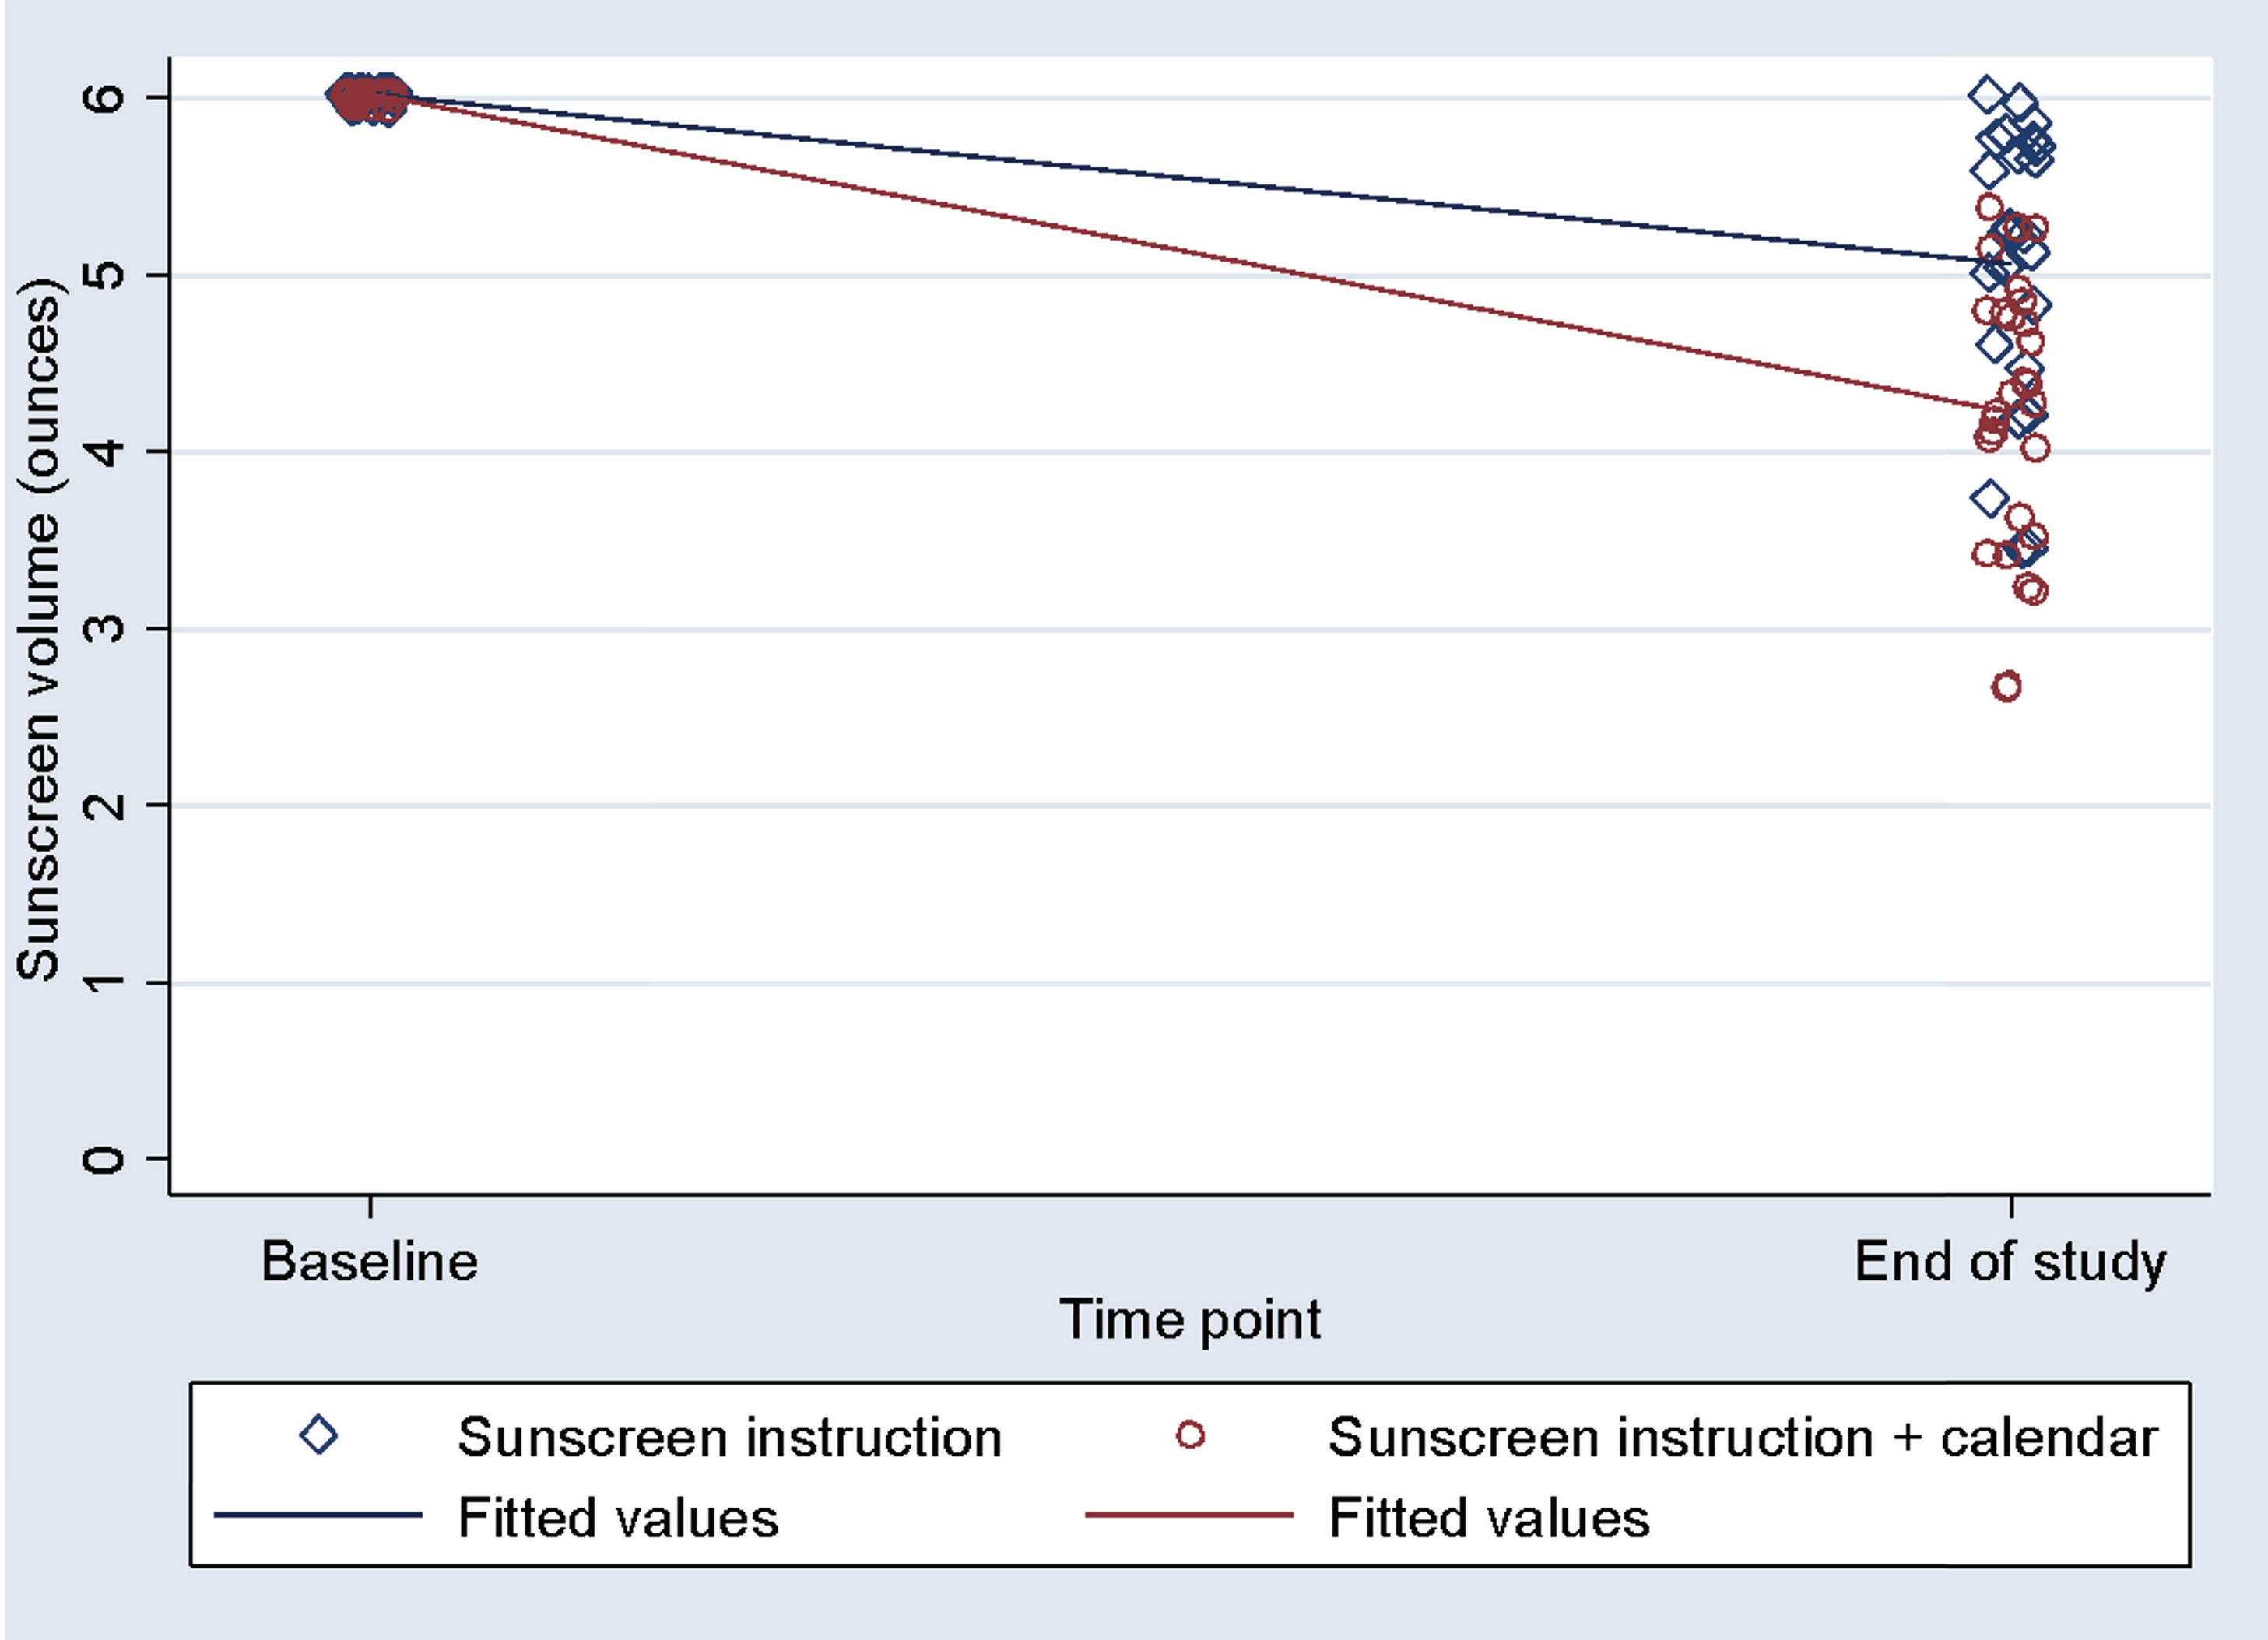
Sunscreen volume (ounces)
0
1
2
3
4
5
6
Baseline
End of study
Time point
Sunscreen instruction
Sunscreen instruction + calendar
Fitted values
Fitted values

Figure 1. Scatterplot of sunscreen volumes at the two evaluation time points by study group (n=53).

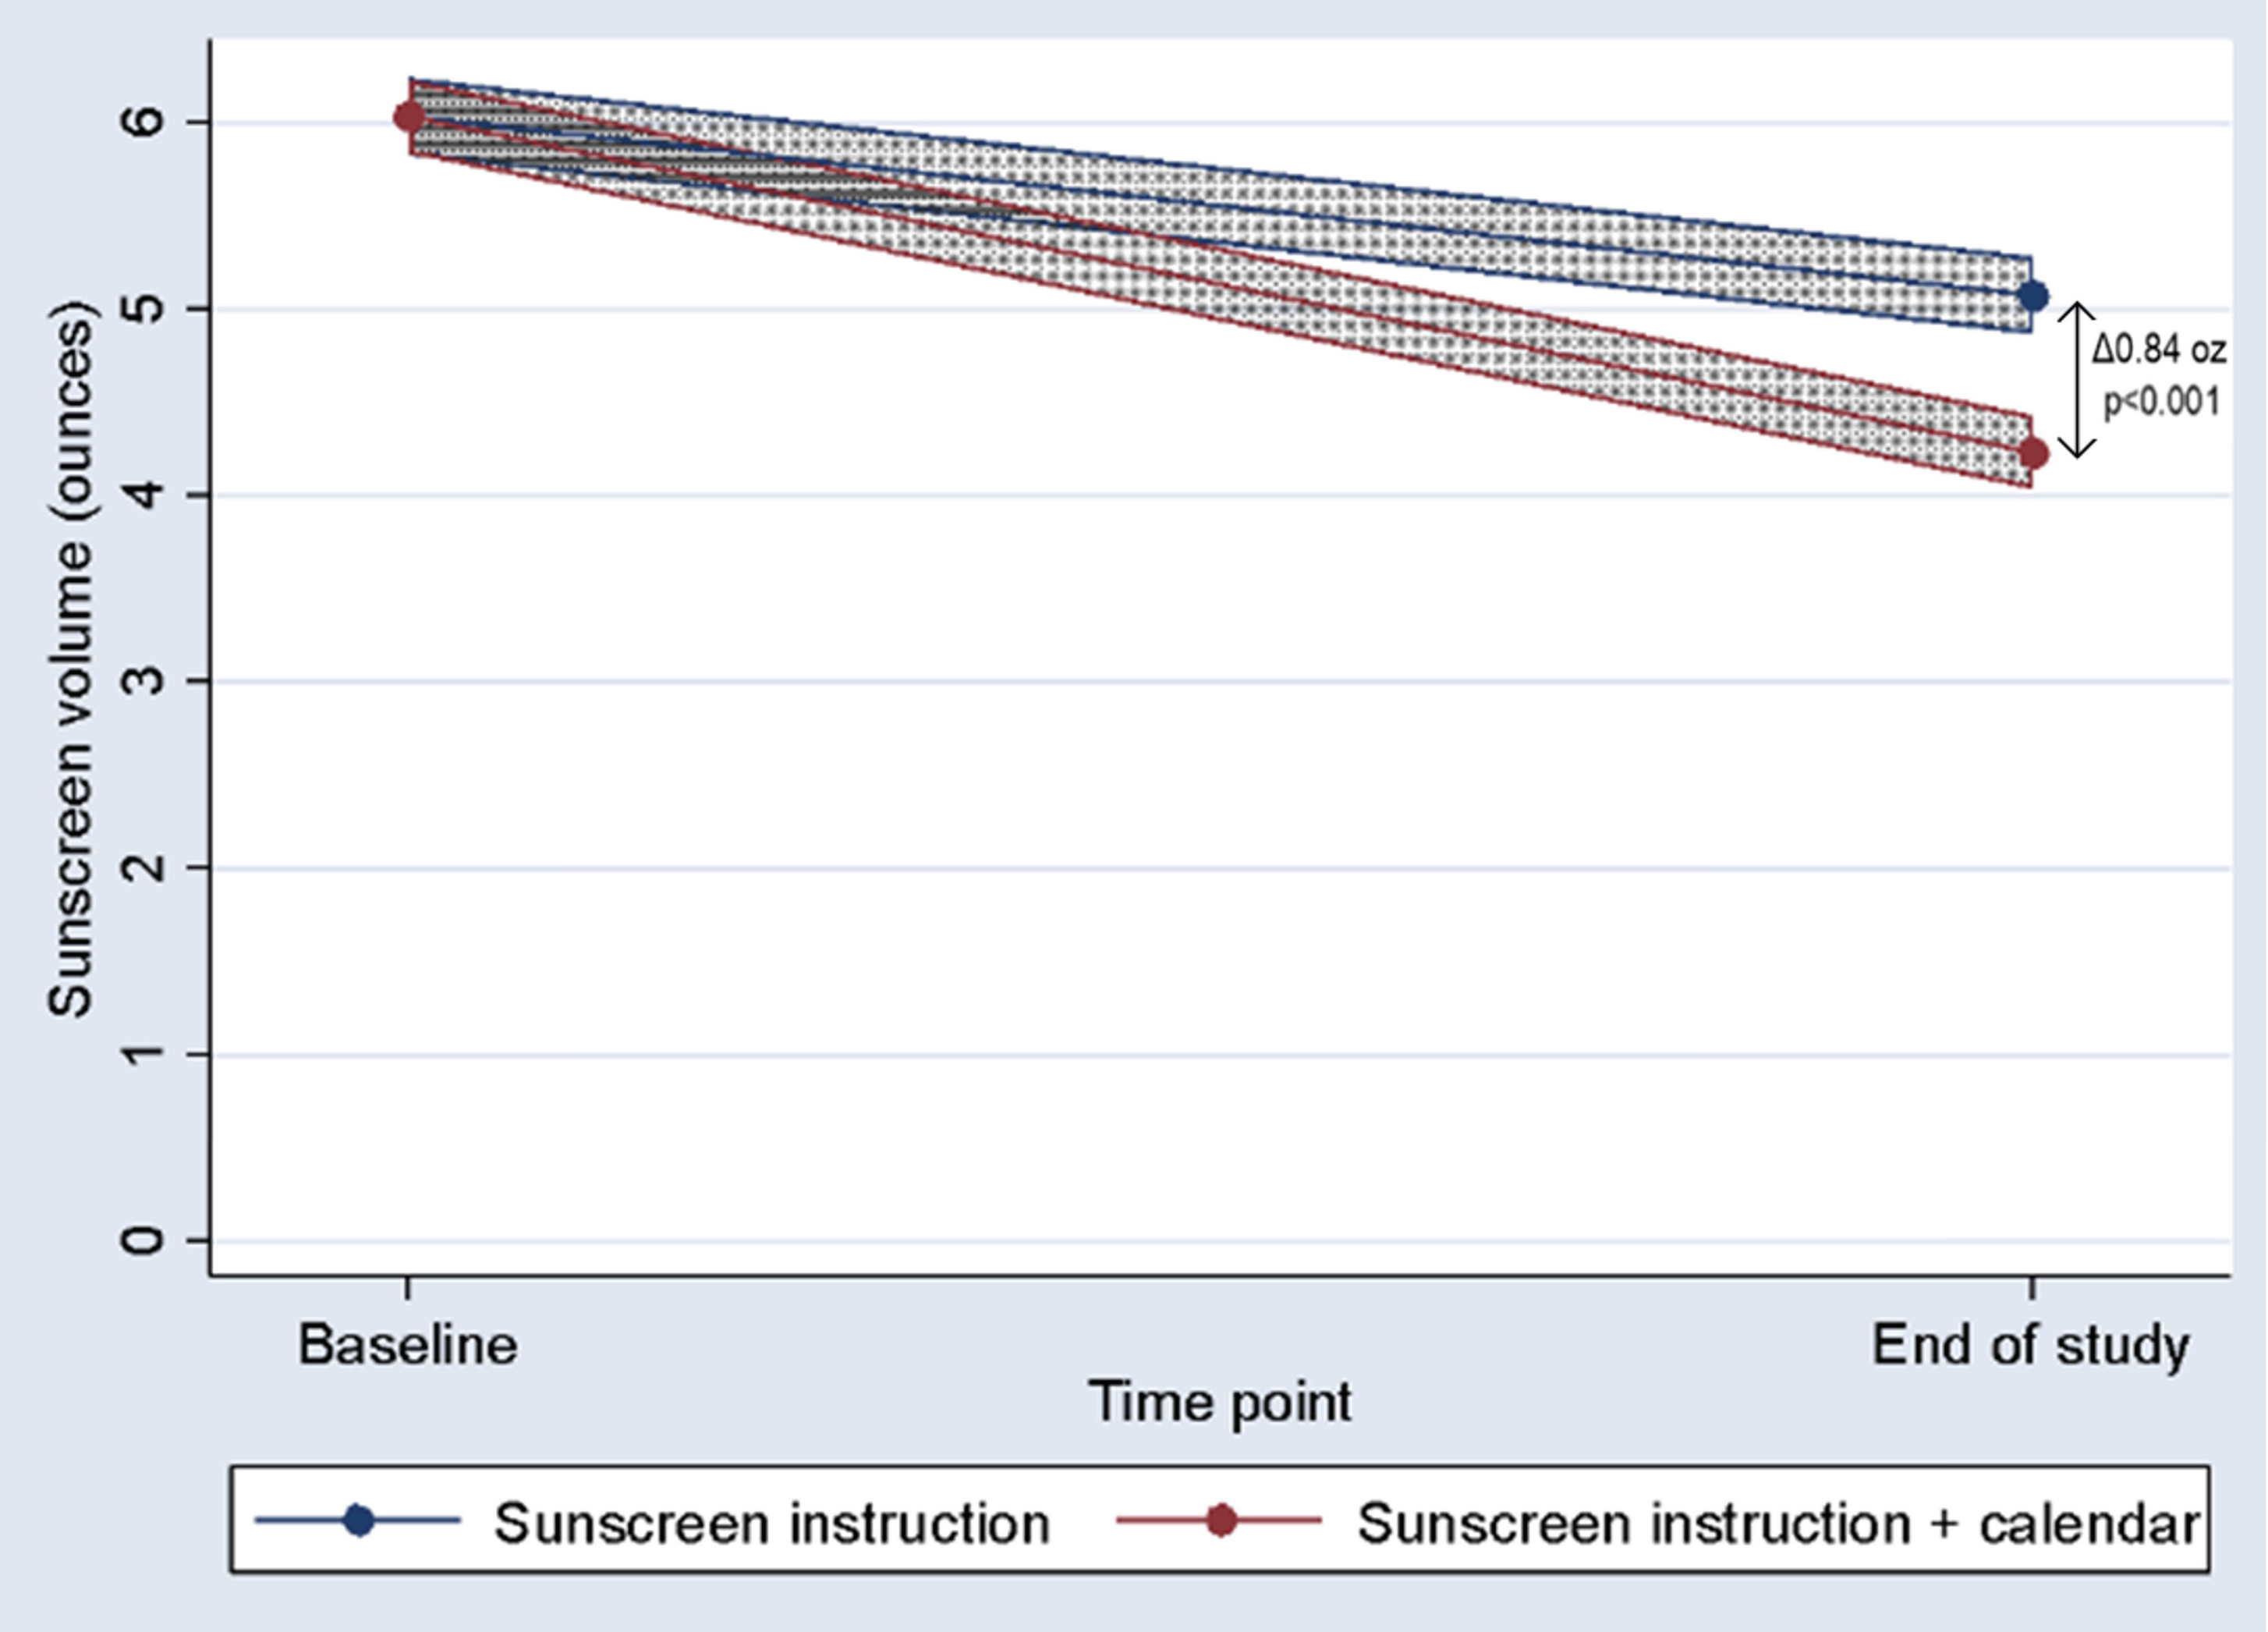
Sunscreen volume (ounces)
0
1
2
3
4
5
6
Δ0.84 oz
p<0.001
Baseline
End of study
Time point
Sunscreen instruction
Sunscreen instruction + calendar

Figure 2. Plot of sunscreen volume trajectories in the two groups by evaluation time point (n=53).

C1 - Internal use

Results

Table 1. Results of sunscreen measurements at baseline and follow-up evaluations along with percent change from baseline.

Group	n	Mean at Baseline	Mean at Follow-Up	Median at Follow-Up	Percent Change From Baseline
Sunscreen Instruction	25	6.03 (SD=0)	5.07 (0.77)	5.13 (IQR=1.1)	-15.96%
Sunscreen Instruction + Calendar	28	6.03 (SD=0)	4.22 (0.75)	4.27 (IQR=1.2)	-29.92%
Overall	53	6.03 (SD=0)	4.62 (0.86)	4.76 (IQR=1.2)	-23.34%

A total of 53 individuals partook in this study. The control group included 25 participants, while the calendar group included 28 participants. Of the 53 total participants, 52 (98%) participants used at least some of the sunscreen over the course of the evaluation period (**Figure 1**). There was no difference in age between the two study arms.

The pre-weight of the sunscreen administered was 6.03 ounces for all participants. The mean and median sunscreen volumes at follow-up were, respectively, 5.07 (0.77) ounces and 5.13 (IQR=1.1) ounces for the control group, 4.22 (0.75) ounces and 4.27 (IQR=1.2) ounces for the calendar group, and 4.62 (0.86) ounces and 4.76 (IQR=1.2) ounces on average (**Table 1**). The control group used 15.96% of the sunscreen, the calendar group used 29.92% of the sunscreen, and 23.34% of the sunscreen was used on average (**Table 1**). A notable difference existed in the volume of sunscreen utilized by participants in each study group, with those in the calendar group applying an additional 0.84 ounces compared to the reference group (p<0.001) (**Figure 2**). These discrepancies were statistically significant across all observed measures (p<0.001).

Discussion

The study demonstrated that integrating calendars as a tracking tool alongside sunscreen application instructions may increase compliance compared to the control group. This suggests that practical strategies like calendar use can reinforce positive health behaviors, such as daily sunscreen application. While further research is needed to validate these findings in larger populations, the results offer valuable insights for designing effective public health interventions to promote sustained sunscreen adherence and reduce sun-related skin damage and diseases.

References

1. Tran T, Song S, Texeira AJ, Rechis R, Nelson KC. Educational interventions to promote sun-protection behaviors in adolescents in the United States: A systematic review. Pediatr Dermatol. 2023;40(4):637-641. doi:10.1111/pde.15335
2. Sim WMB, Zeng MX, Rojas-Garcia A. The effectiveness of educational programmes in promoting sun protection among children under the age of 18: a systematic review and meta-analysis. J Eur Acad Dermatol Venereol. 2021;35(11):2154-2165. doi:10.1111/jdv.17599
3. Chambergo-Michilot D, Tellez WA, Becerra-Chauca N, Zafra-Tanaka JH, Taype-Rondan A. Text message reminders for improving sun protection habits: A systematic review. PLoS One. 2020;15(5):e0233220. Published 2020 May 19. doi:10.1371/journal.pone.0233220
4. Horsham C, Auster J, Sendall MC, et al. Interventions to decrease skin cancer risk in outdoor workers: update to a 2007 systematic review. BMC Res Notes. 2014;7:10. Published 2014 Jan 7. doi:10.1186/1756-0500-7-10